The stapled peptide ALRN-6924, a dual inhibitor of MDMX and MDM2, enhances antitumor efficacy of paclitaxel and Nab-paclitaxel in TP53 wild-type breast cancer models

Seyed Pairawan[1], Erkan Yuca[1], Kurt Evans[1], Allen Annis[2], Ming Zhao[1] Narayana Narasimhan[2], David Sutton[2], Luis Carvajal[2], Jian-Guo Ren[2], Solimar Santiago[2], Vincent Guerlavais[2], Argun Akcakanat[1], Coya Tapia[1], Ecaterina Dumbrava[1], Manuel Aivado[2], Funda Meric-Bernstam[1]

[1] The University of Texas MD Anderson Cancer Center, Houston, TX, [2] Aileron Therapeutics, Watertown, MA

T32 CA009599

Background

MDMX and MDM2 are endogenous inhibitors of the p53 tumor suppressor protein. MDMX levels are frequently elevated in luminal breast cancer, which generally express wild-type (wt) p53. ALRN-6924, an α-helical stapled p53 peptide, is the first and only dual inhibitor of MDMX and MDM2 currently in clinical trials for solid tumors and hematological malignancies. We sought to determine the antitumor efficacy of the combination of ALRN-6924 with taxanes in wild-type p53 estrogen receptor positive (ER+) models of human breast cancer.

Methods

Sulforhodamine B (SRB) colorimetric assay was used to assess the cytotoxicity of the combination of ALRN-6924 with taxanes *in vitro*. Athymic nude mice were implanted with wt p53 and ER+ MCF-7 and ZR-75-1 cells and treated for four weeks with ALRN-6924 alone and in combination with paclitaxel in cremaphor (Taxol®, study #1) or a nanoparticle-albumin-bound (nab) formulation (Abraxane®, study #2). In study #1, ALRN-6924 (5, 10 mg/kg) was dosed twice weekly and paclitaxel (10, 15 mg/kg) was dosed weekly, with paclitaxel administered 6 h prior to ALRN-6924. In study #2, ALRN-6924 alone (5 mg/kg) was dosed twice weekly while nab-paclitaxel (15 mg/kg) was administered weekly in combination at -24h, -6h, 0h, +6h, or +24h relative to ALRN-6924 administration.

Results

Cell line	ALRN-6924	Paclitaxel
MCF-7	117 nM	0.03 nM
ZR-75-1	606 nM	0.2 nM

Table 1. Sensitivity of hormone positive breast cancer cells to ALRN-6924 and Paclitaxel. Cells were seeded in 96-well plates and treated with individual drugs at various dose ranges for 3 days. The cells survival was measured from optical density (OD) value after SRB staining. IC50s were calculated from dose curves using Calcusyn.

Results

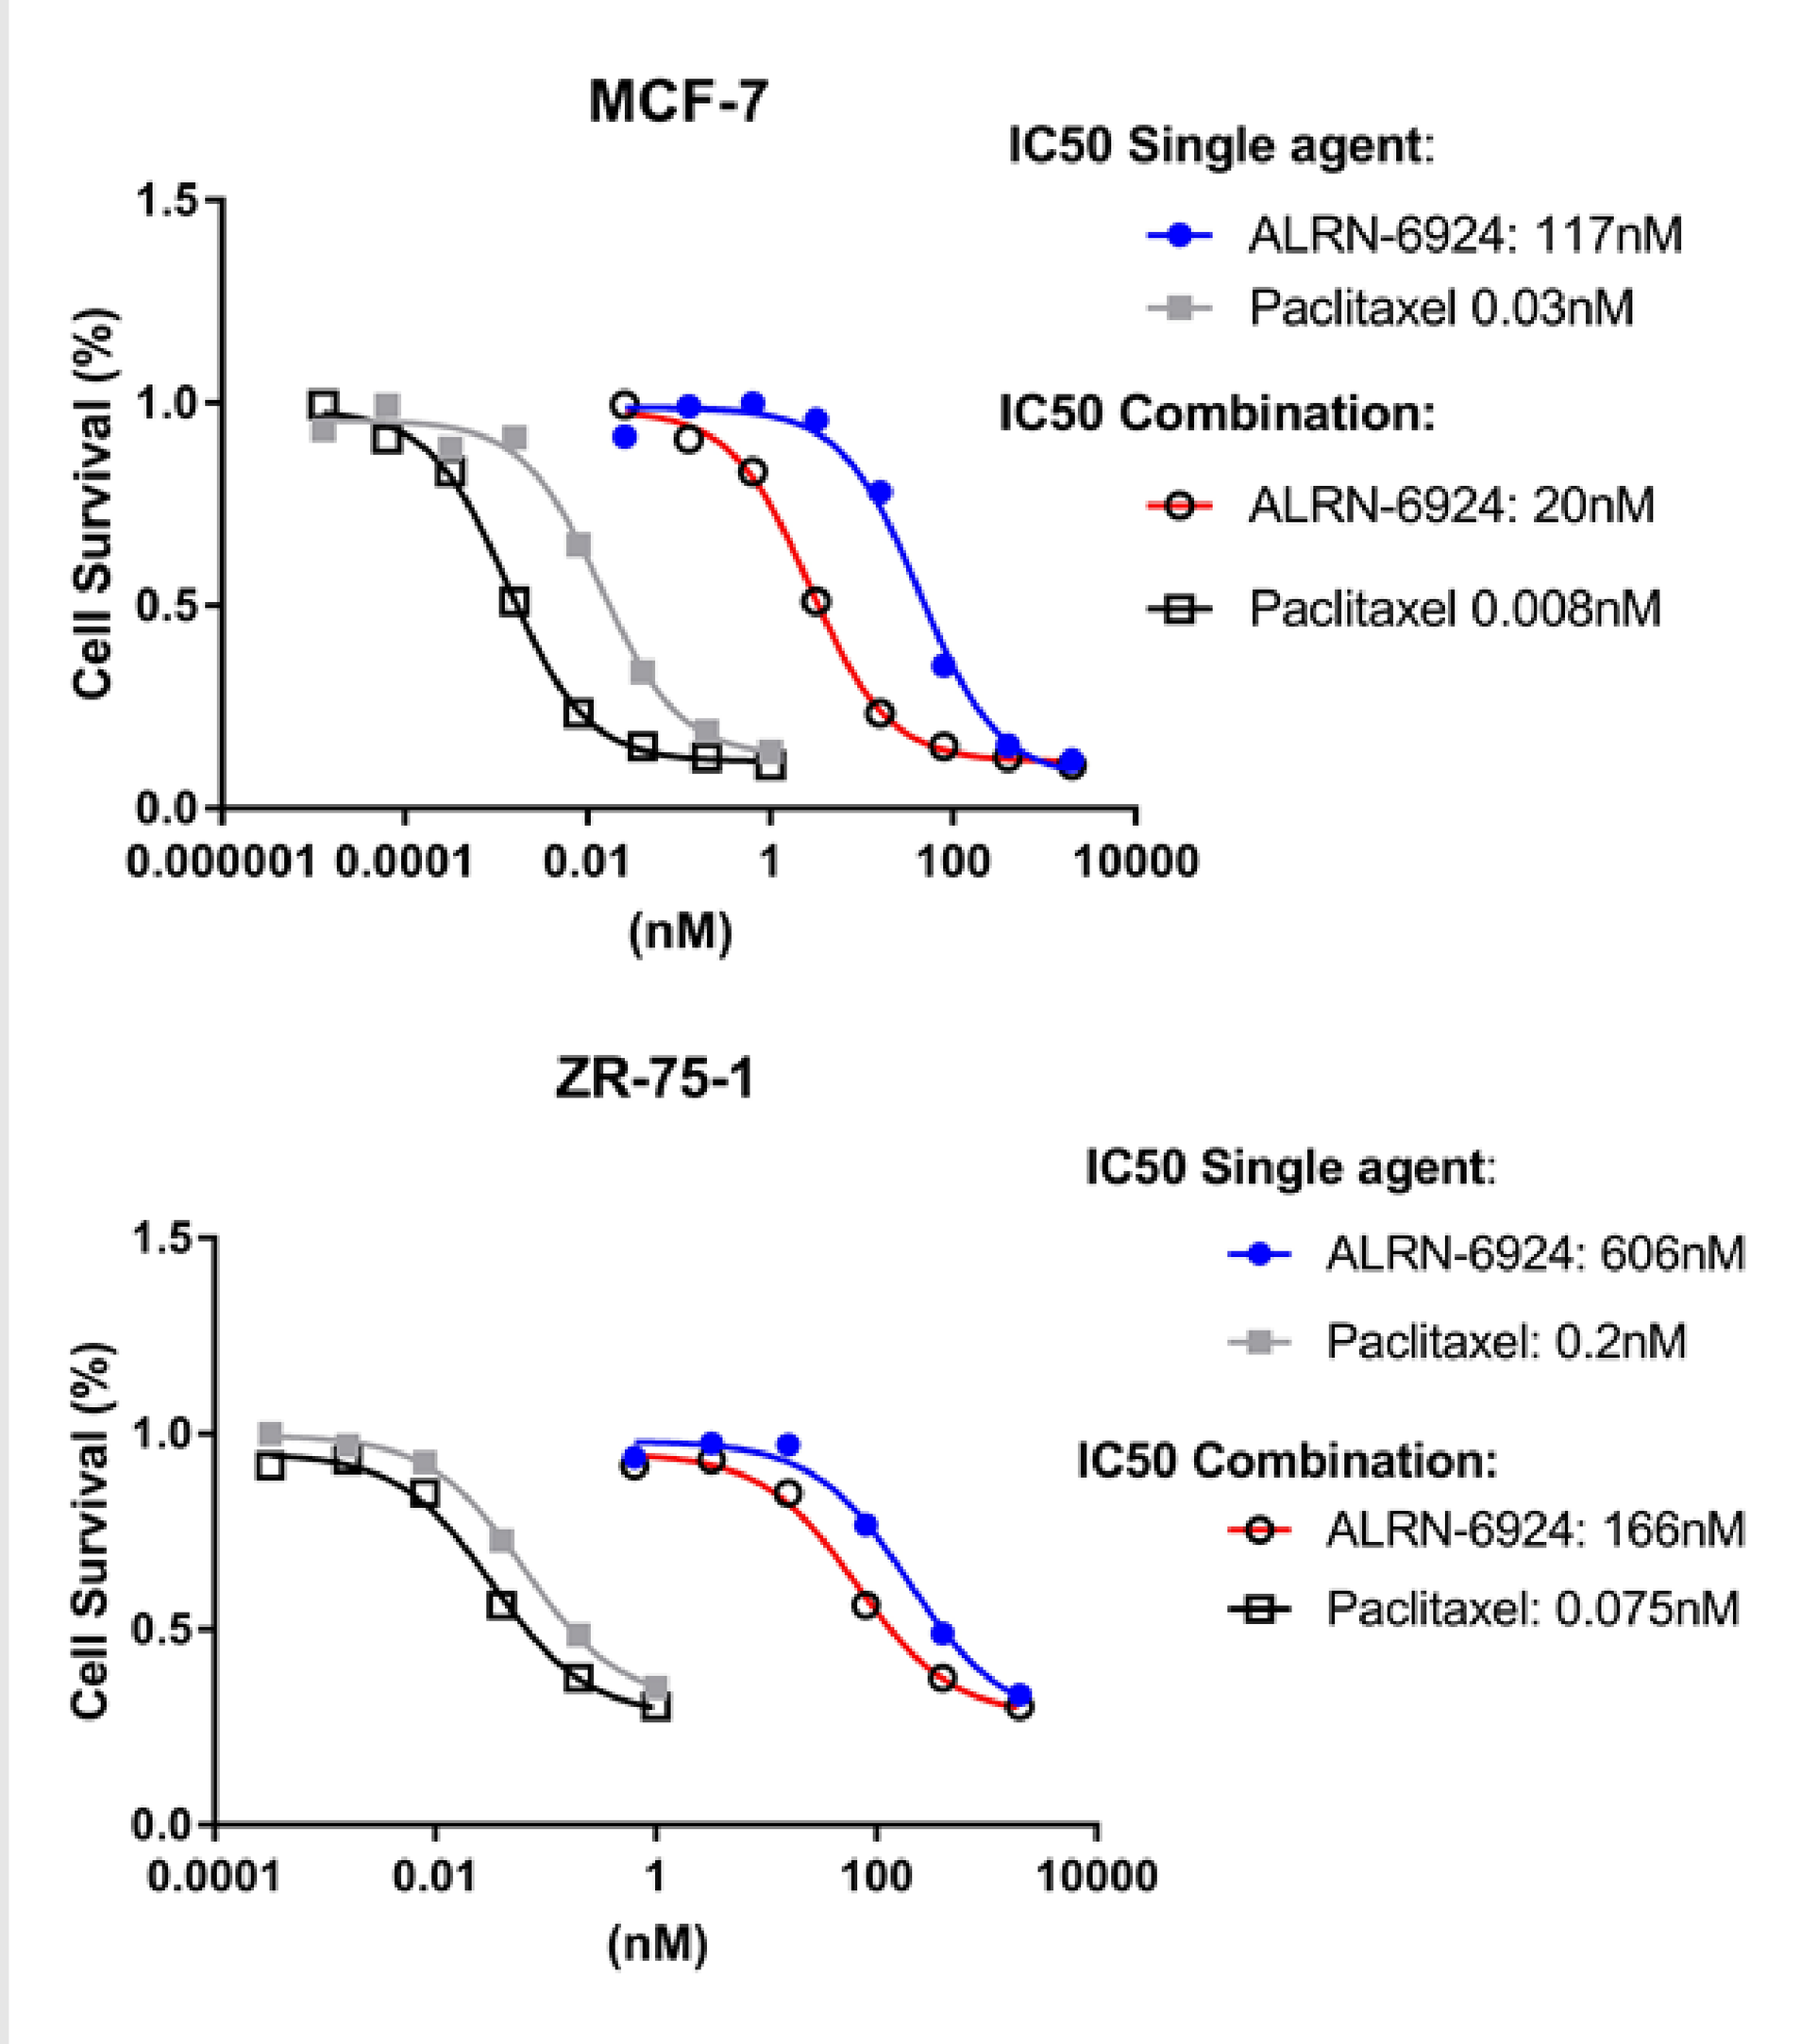
MCF-7
IC50 Single agent:
ALRN-6924: 117nM
Paclitaxel 0.03nM
IC50 Combination:
ALRN-6924: 20nM
Paclitaxel 0.008nM
Cell Survival (%)
(nM)
ZR-75-1
IC50 Single agent:
ALRN-6924: 606nM
Paclitaxel: 0.2nM
IC50 Combination:
ALRN-6924: 166nM
Paclitaxel: 0.075nM

Figure 1. The combination of ALRN-6924 and Paclitaxel demonstrates synergistic activity on cell survival in MCF-7 and ZR-75-1 breast cancer cells. Cells seeded in 96-well plates were incubated with ALRN-6924 and Paclitaxel at various dose range ratios, along with single drug treatment. IC50s were obtained from each dose curves using Calcusyn.

Cell line	ALRN-6924 + Paclitaxel combination indices
MCF-7	0.44
ZR-75-1	0.52

Table 2. Combination indices across wt p53 hormone positive breast cancer cells to ALRN-6924 and Paclitaxel. Combination index (CI) was calculated using Calcusyn. CI < 1: Synergistic, CI=1 Additive, CI > 1 Antagonistic effect respectively.

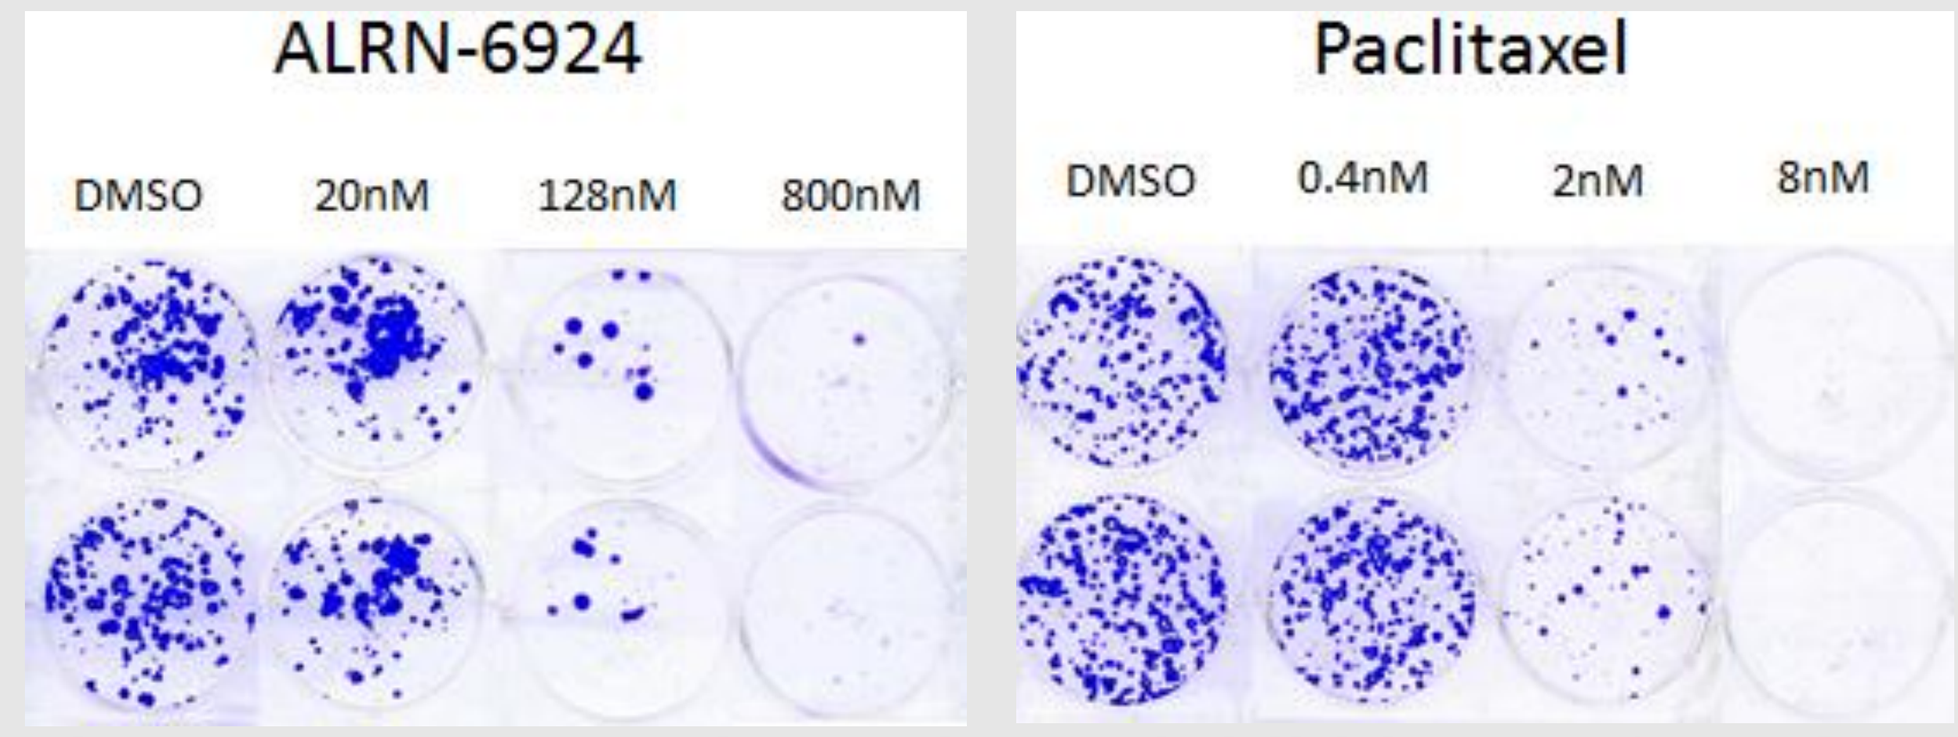
ALRN-6924
DMSO
20nM
128nM
800nM
Paclitaxel
DMSO
0.4nM
2nM
8nM

Figure 2. Dose-dependent inhibition of colony formation of MCF-7 cells by ALRN-6924 and Paclitaxel. Cells seeded in 6-well plates were treated with drugs at various doses for 10 days followed by fixation of cells and Crystal Violet staining.

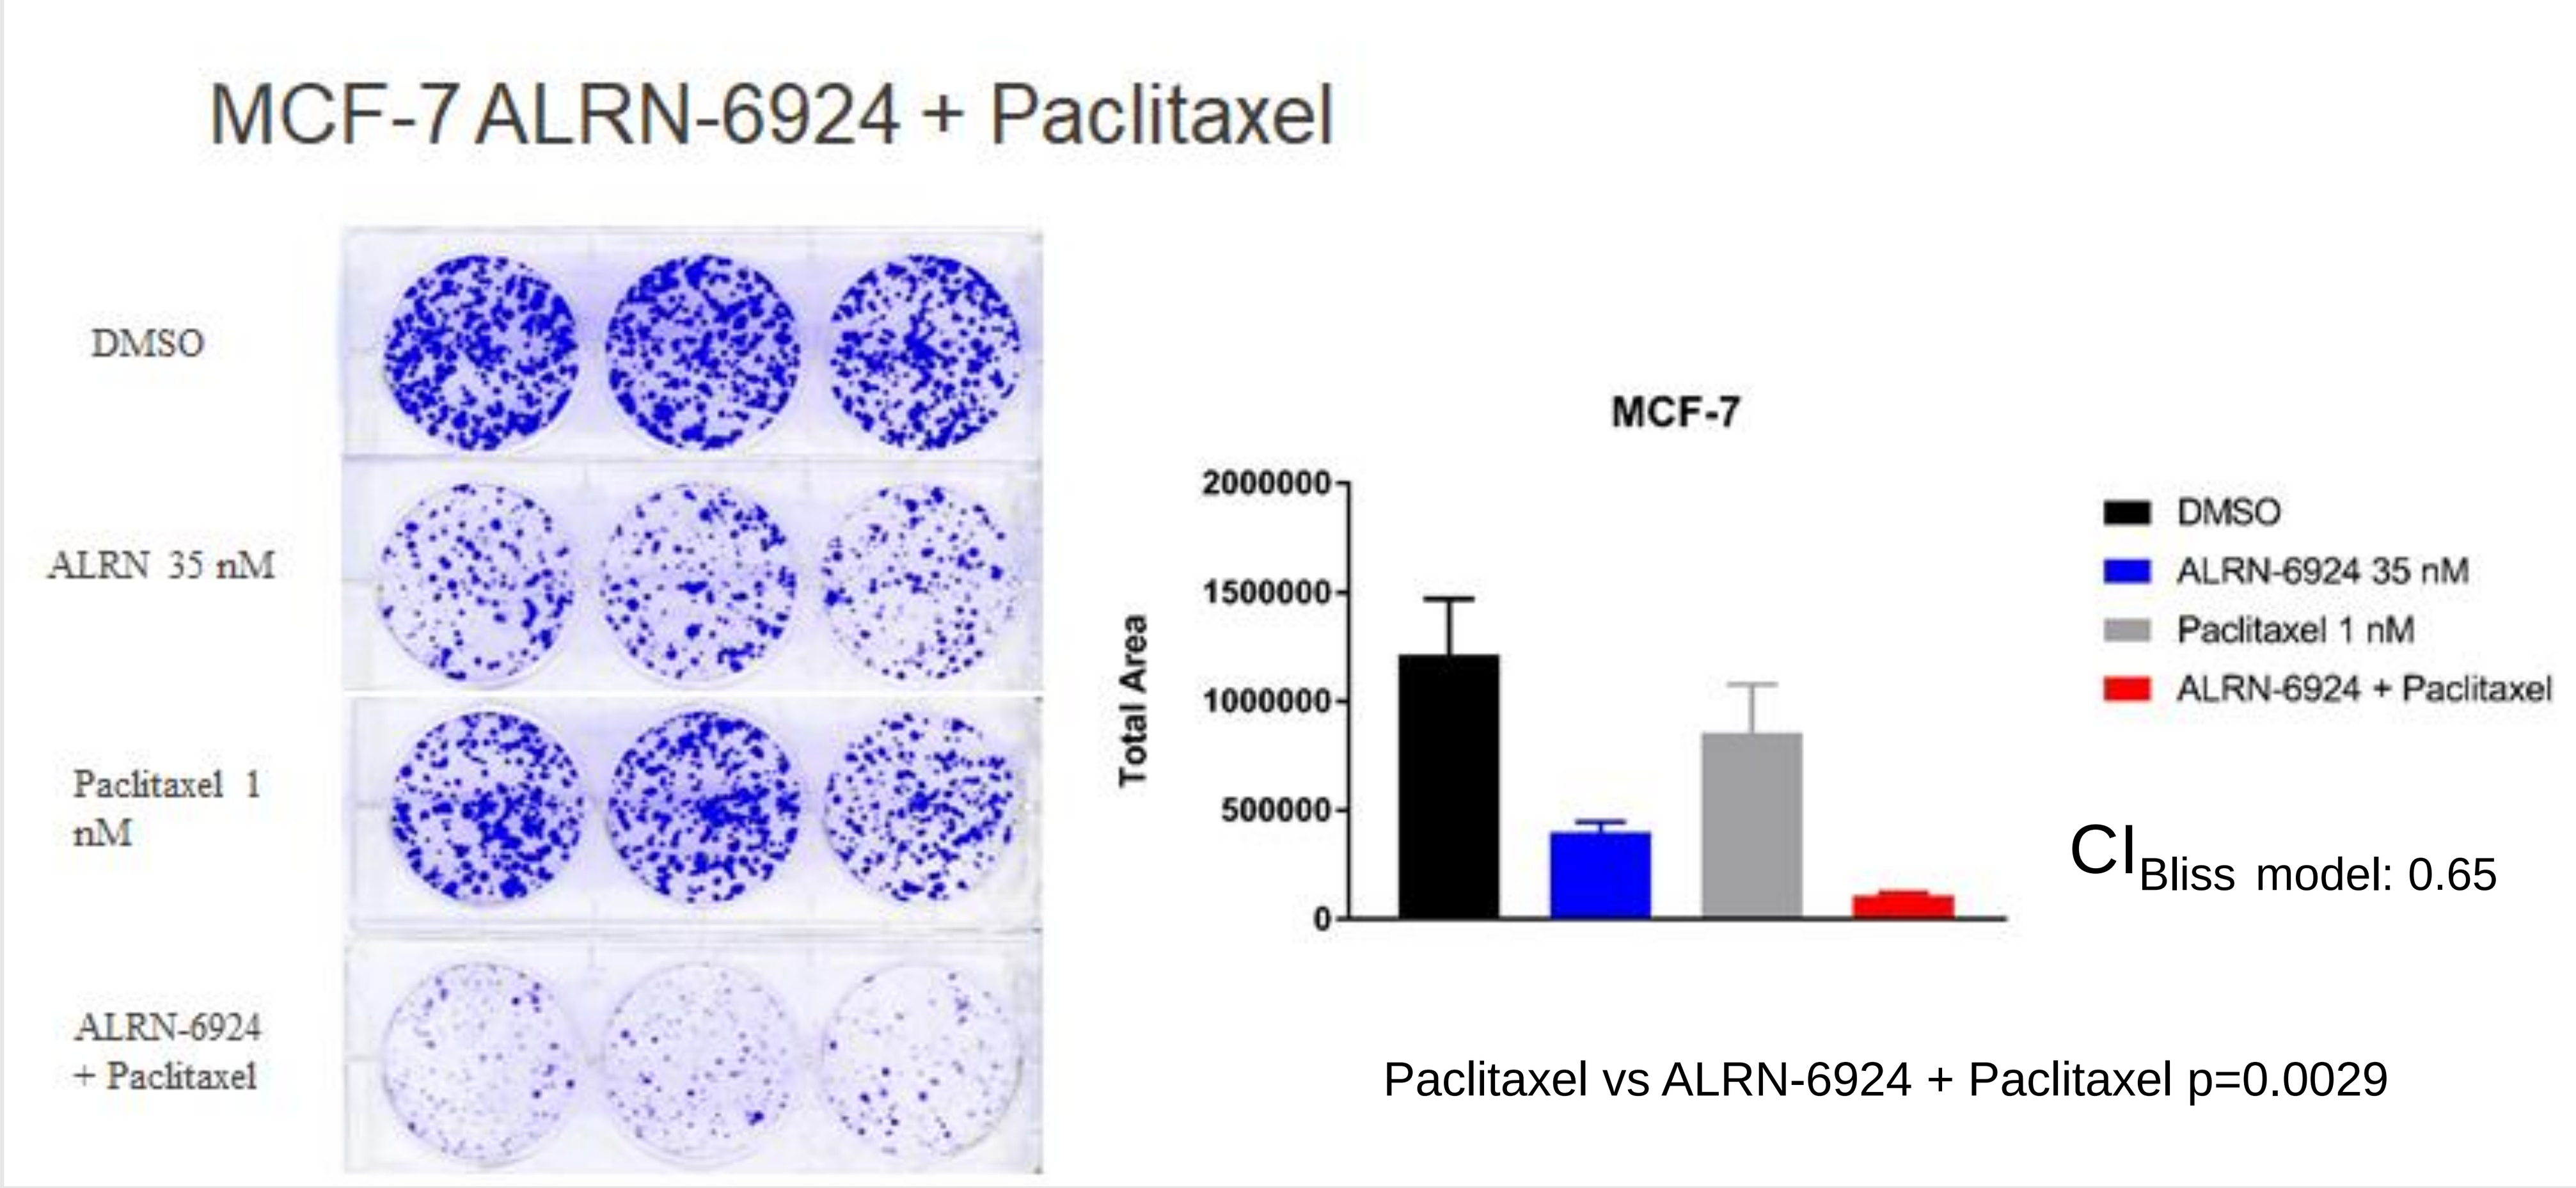
MCF-7 ALRN-6924 + Paclitaxel
DMSO
ALRN 35 nM
Paclitaxel 1 nM
ALRN-6924 + Paclitaxel
MCF-7
Total Area
DMSO
ALRN-6924 35 nM
Paclitaxel 1 nM
ALRN-6924 + Paclitaxel
$CI_{Bliss\ model}$: 0.65
Paclitaxel vs ALRN-6924 + Paclitaxel p=0.0029

Figure 3. **Combining ALRN-6924 and Paclitaxel demonstrates synergistic activity on colony formation assay of MCF-7 cells**. Cells were treated with single or combined drugs at the indicated doses for 10 days. Total colony area was quantitated by ImageJ. Combination index was calculated using Bliss model (CI<1: synergistic; CI=1: additive; CI>1: antagonistic effect respectively)

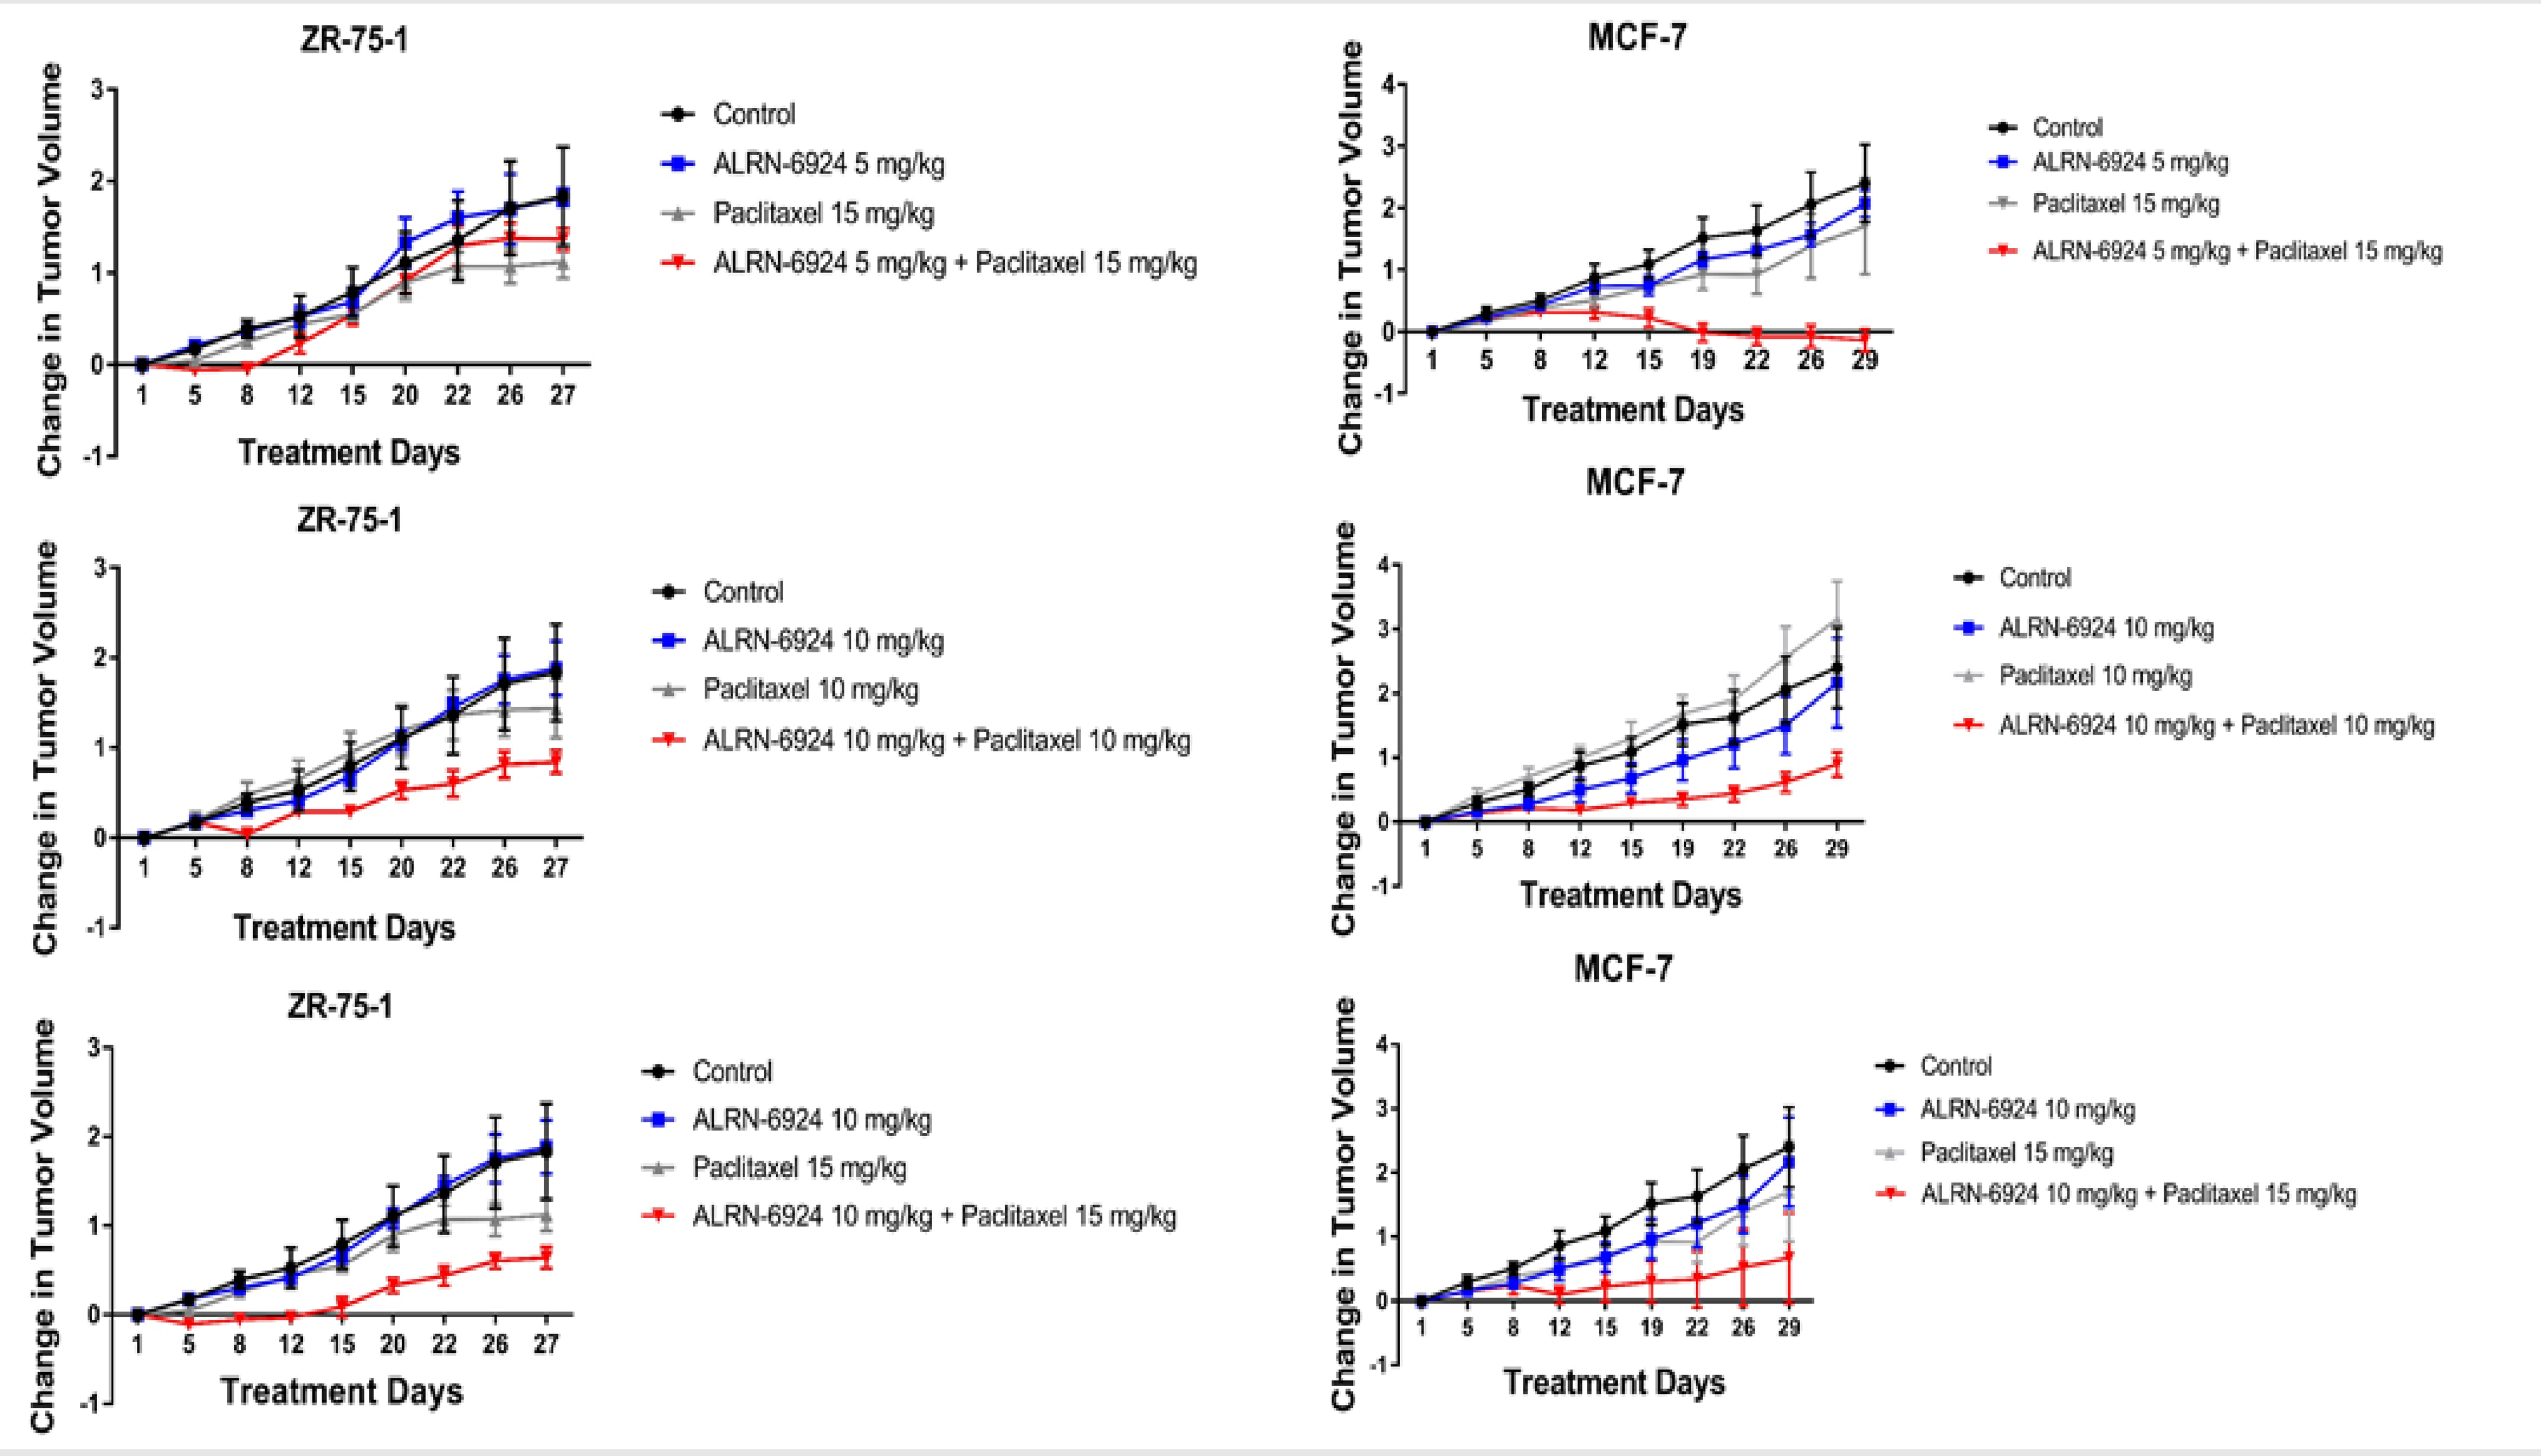
ZR-75-1
Control
ALRN-6924 5 mg/kg
Paclitaxel 15 mg/kg
ALRN-6924 5 mg/kg + Paclitaxel 15 mg/kg
ZR-75-1
Control
ALRN-6924 10 mg/kg
Paclitaxel 10 mg/kg
ALRN-6924 10 mg/kg + Paclitaxel 10 mg/kg
ZR-75-1
Control
ALRN-6924 10 mg/kg
Paclitaxel 15 mg/kg
ALRN-6924 10 mg/kg + Paclitaxel 15 mg/kg
MCF-7
MCF-7
MCF-7
Change in Tumor Volume
Treatment Days

Figure 4. Growth inhibition of breast cancer tumors by ALRN-6924 and Paclitaxel in mice. The combination of ALRN-6924 and paclitaxel significantly inhibited MCF-7 tumor growth compared to either agent alone ($p \leq 0.005$). Paclitaxel 15 mg/kg + ALRN-6924 5 mg/kg in MCF-7 resulted in the greatest tumor inhibition with average tumor size decreased by 13% at four weeks versus the starting size.

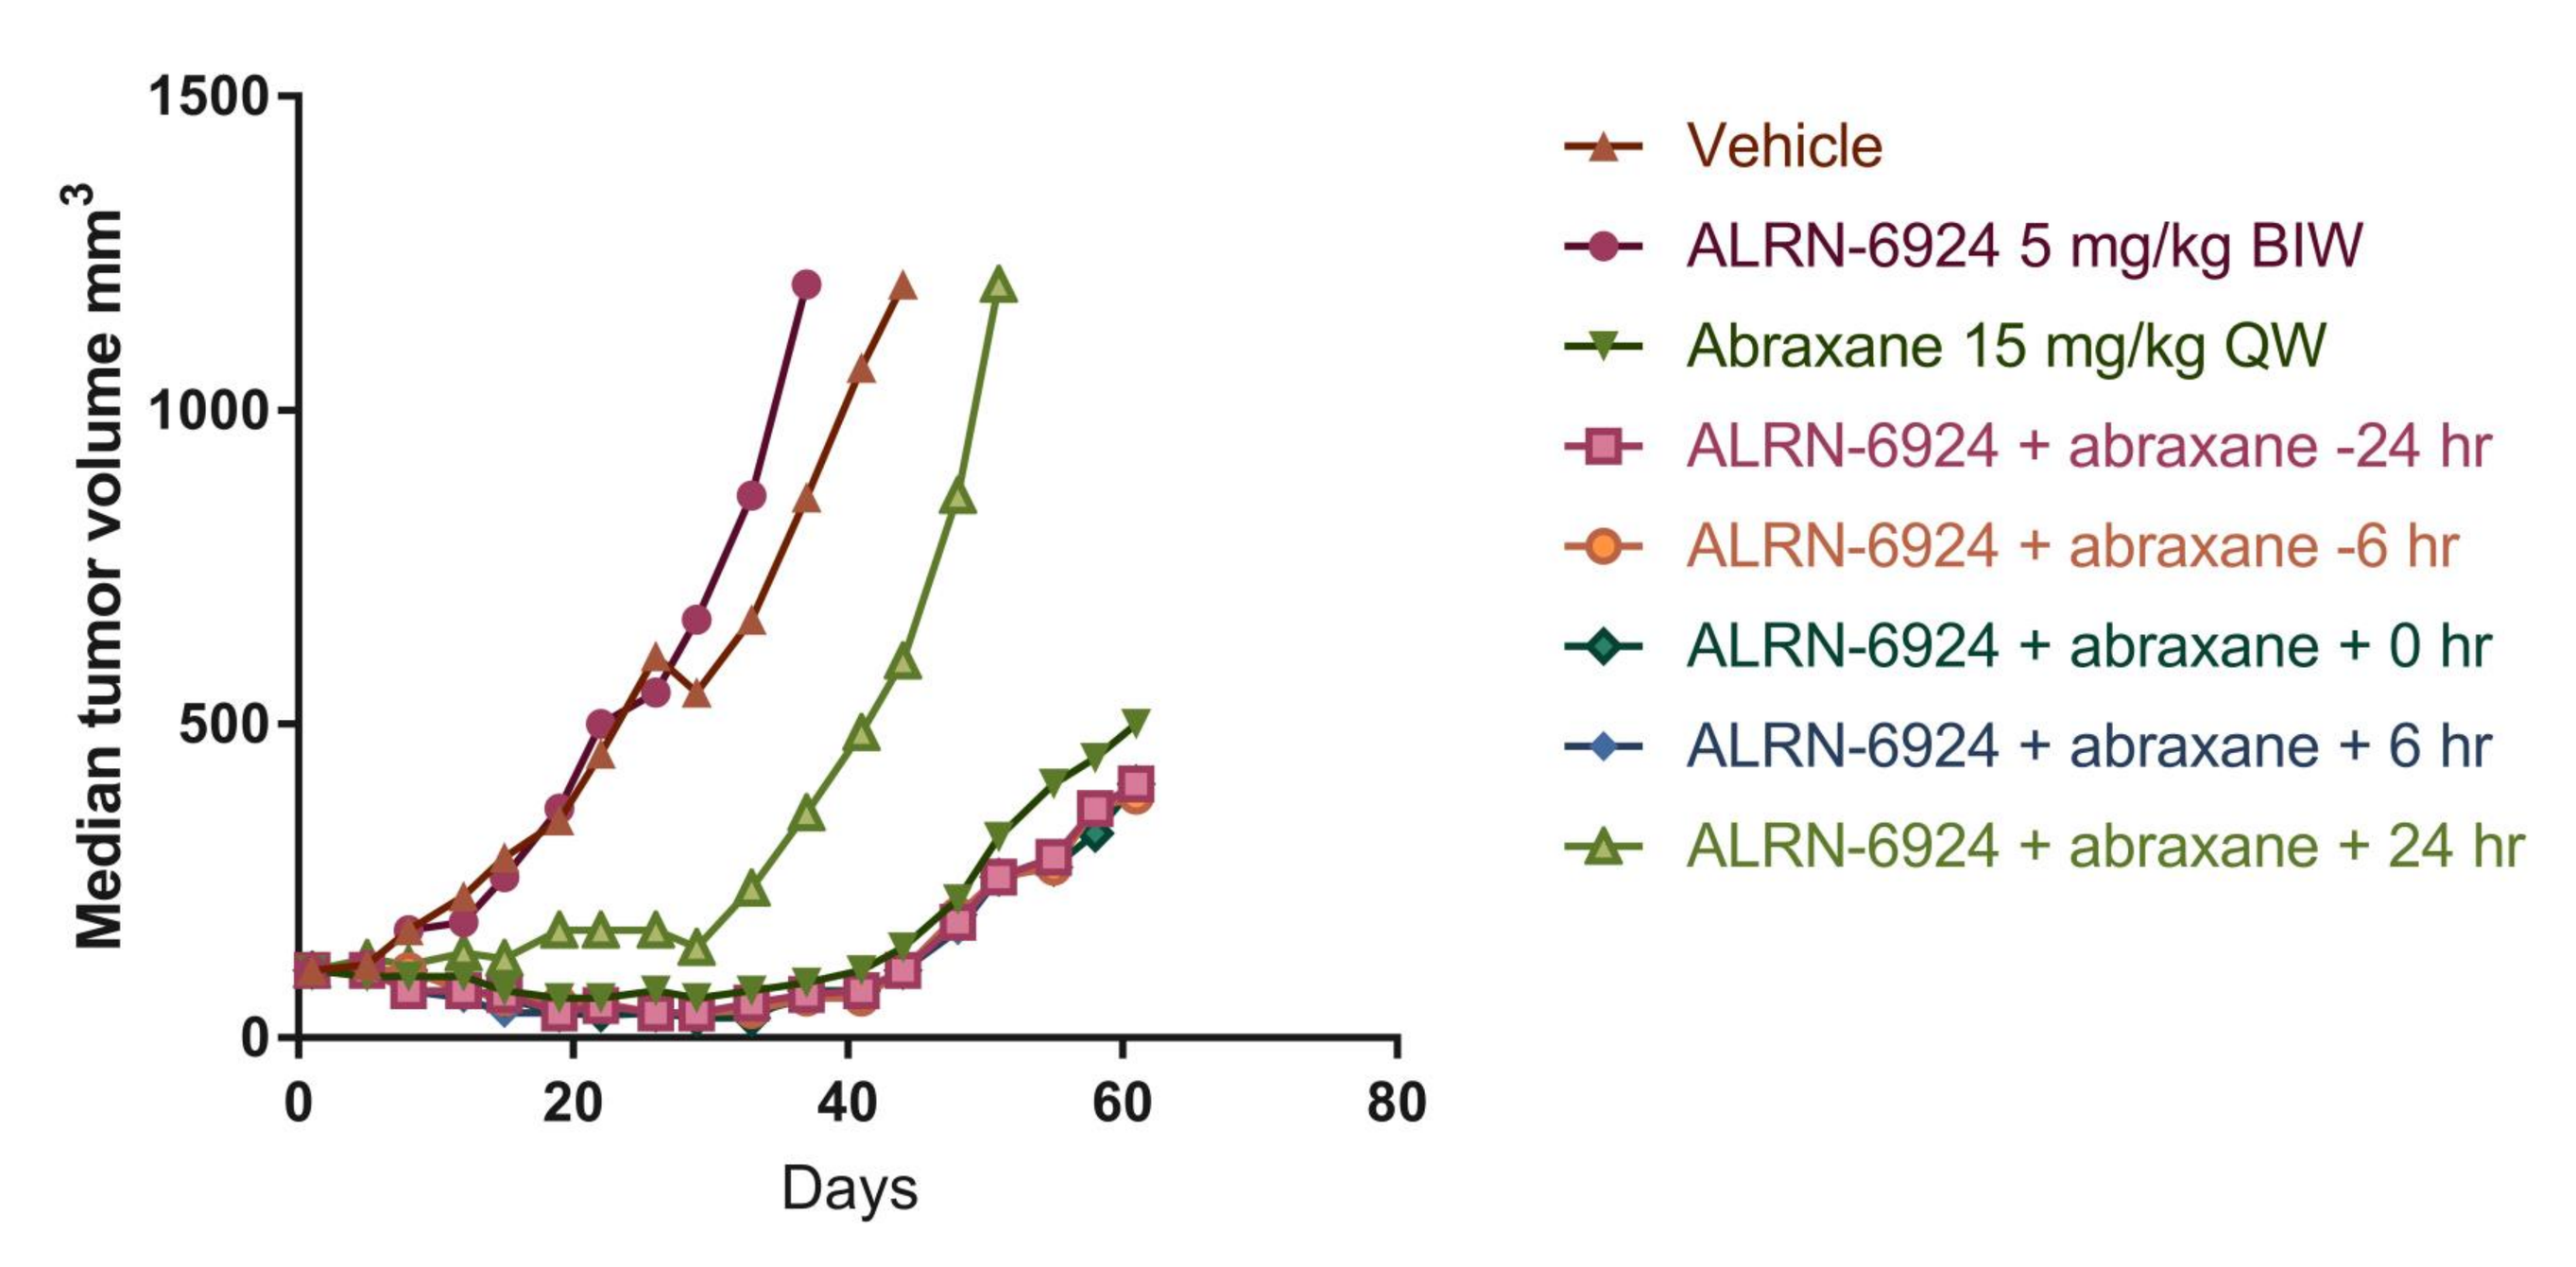
Median tumor volume mm3
Days
Vehicle
ALRN-6924 5 mg/kg BIW
Abraxane 15 mg/kg QW
ALRN-6924 + abraxane -24 hr
ALRN-6924 + abraxane -6 hr
ALRN-6924 + abraxane + 0 hr
ALRN-6924 + abraxane + 6 hr
ALRN-6924 + abraxane + 24 hr

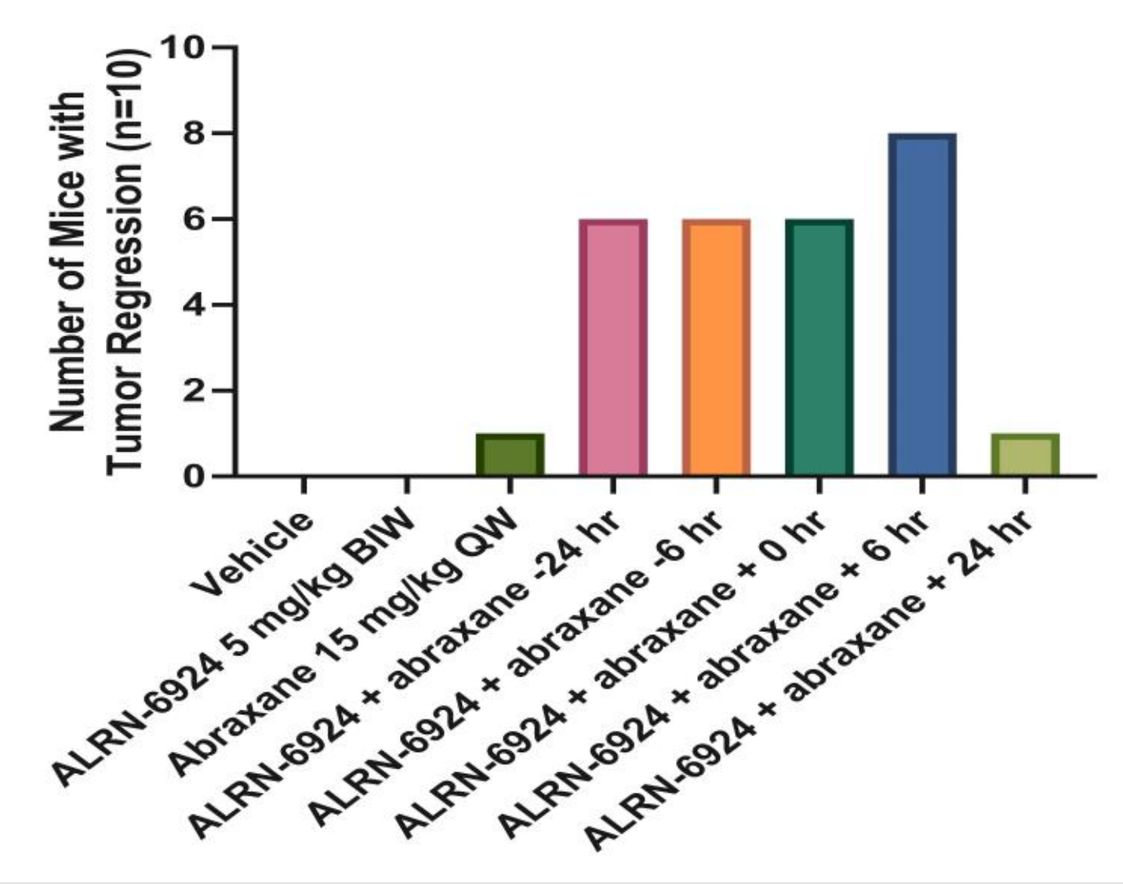
Number of Mice with Tumor Regression (n=10)
Vehicle
ALRN-6924 5 mg/kg BIW
Abraxane 15 mg/kg QW
ALRN-6924 + abraxane -24 hr
ALRN-6924 + abraxane -6 hr
ALRN-6924 + abraxane + 0 hr
ALRN-6924 + abraxane + 6 hr
ALRN-6924 + abraxane + 24 hr

6924	0/10
Abraxane alone	1PR/10
6924 + Abraxane 0h	6PR/10
6924 + Abraxane +6h	8PR/10
6924 + Abraxane -6h	6PR/10
6924 + Abraxane -24h	6PR/10
6924 + Abraxane +24h	1CR/0PR/10

Figure 5 and 6. **The combination of Nab-Paclitaxel with ALRN-6924 in MCF-7 xenograft models resulted in improved antitumor efficacy over either single agent.** ALRN-6924 was administered -6h to +24h relative to nab-paclitaxel. significant increase in the number of tumor regressions (up to 6/10 with 3 consecutive measurements <50% of starting volume) compared to nab-paclitaxel alone (1/10, p<0.005). When ALRN-6924 was administered 24h prior to nab-paclitaxel, there was a marked decrease in efficacy and no tumor regressions were observed

Conclusion

1. There is a significant increase in antitumor efficacy observed with ALRN-6924 in combination with paclitaxel in preclinical breast cancer tumor models.
2. Combining ALRN-6924 with nab-paclitaxel resulted in significant increase in number of tumor regressions.
3. These findings support further evaluation of described combinations in patients with breast cancer.

Funding

This work was funded by the T32 grant (CA009599) , Aileron Therapeutics and Nellie B. Connally Breast Cancer Endowment.